BDB001, an intravenously administered Toll-Like Receptor 7 and 8 (TLR7/8) agonist, in combination with pembrolizumab in advanced solid tumors: Phase 1 safety and efficacy results

Manish R. Patel[1,4], Anthony W. Tolcher[2] Drew W. Rasco[3], Melissa L. Johnson[4], Angela T. Alistar[5], Lixin Li[6], Alexander H. Chung[6], Robert H.I. Andtbacka[6]

[1]Florida Cancer Specialists & Research Institute [2]NEXT Oncology [3]South Texas Accelerated Research Therapeutics [4]Sarah Cannon Research Institute [5]Atlantic Health System, Morristown Medical Center [6]Seven and Eight Biopharmaceuticals Inc.

2512

Seven&Eight Biopharma

Background / Study Design

BDB001 is an immune modulator capable of generating both innate and adaptive immunity against tumors

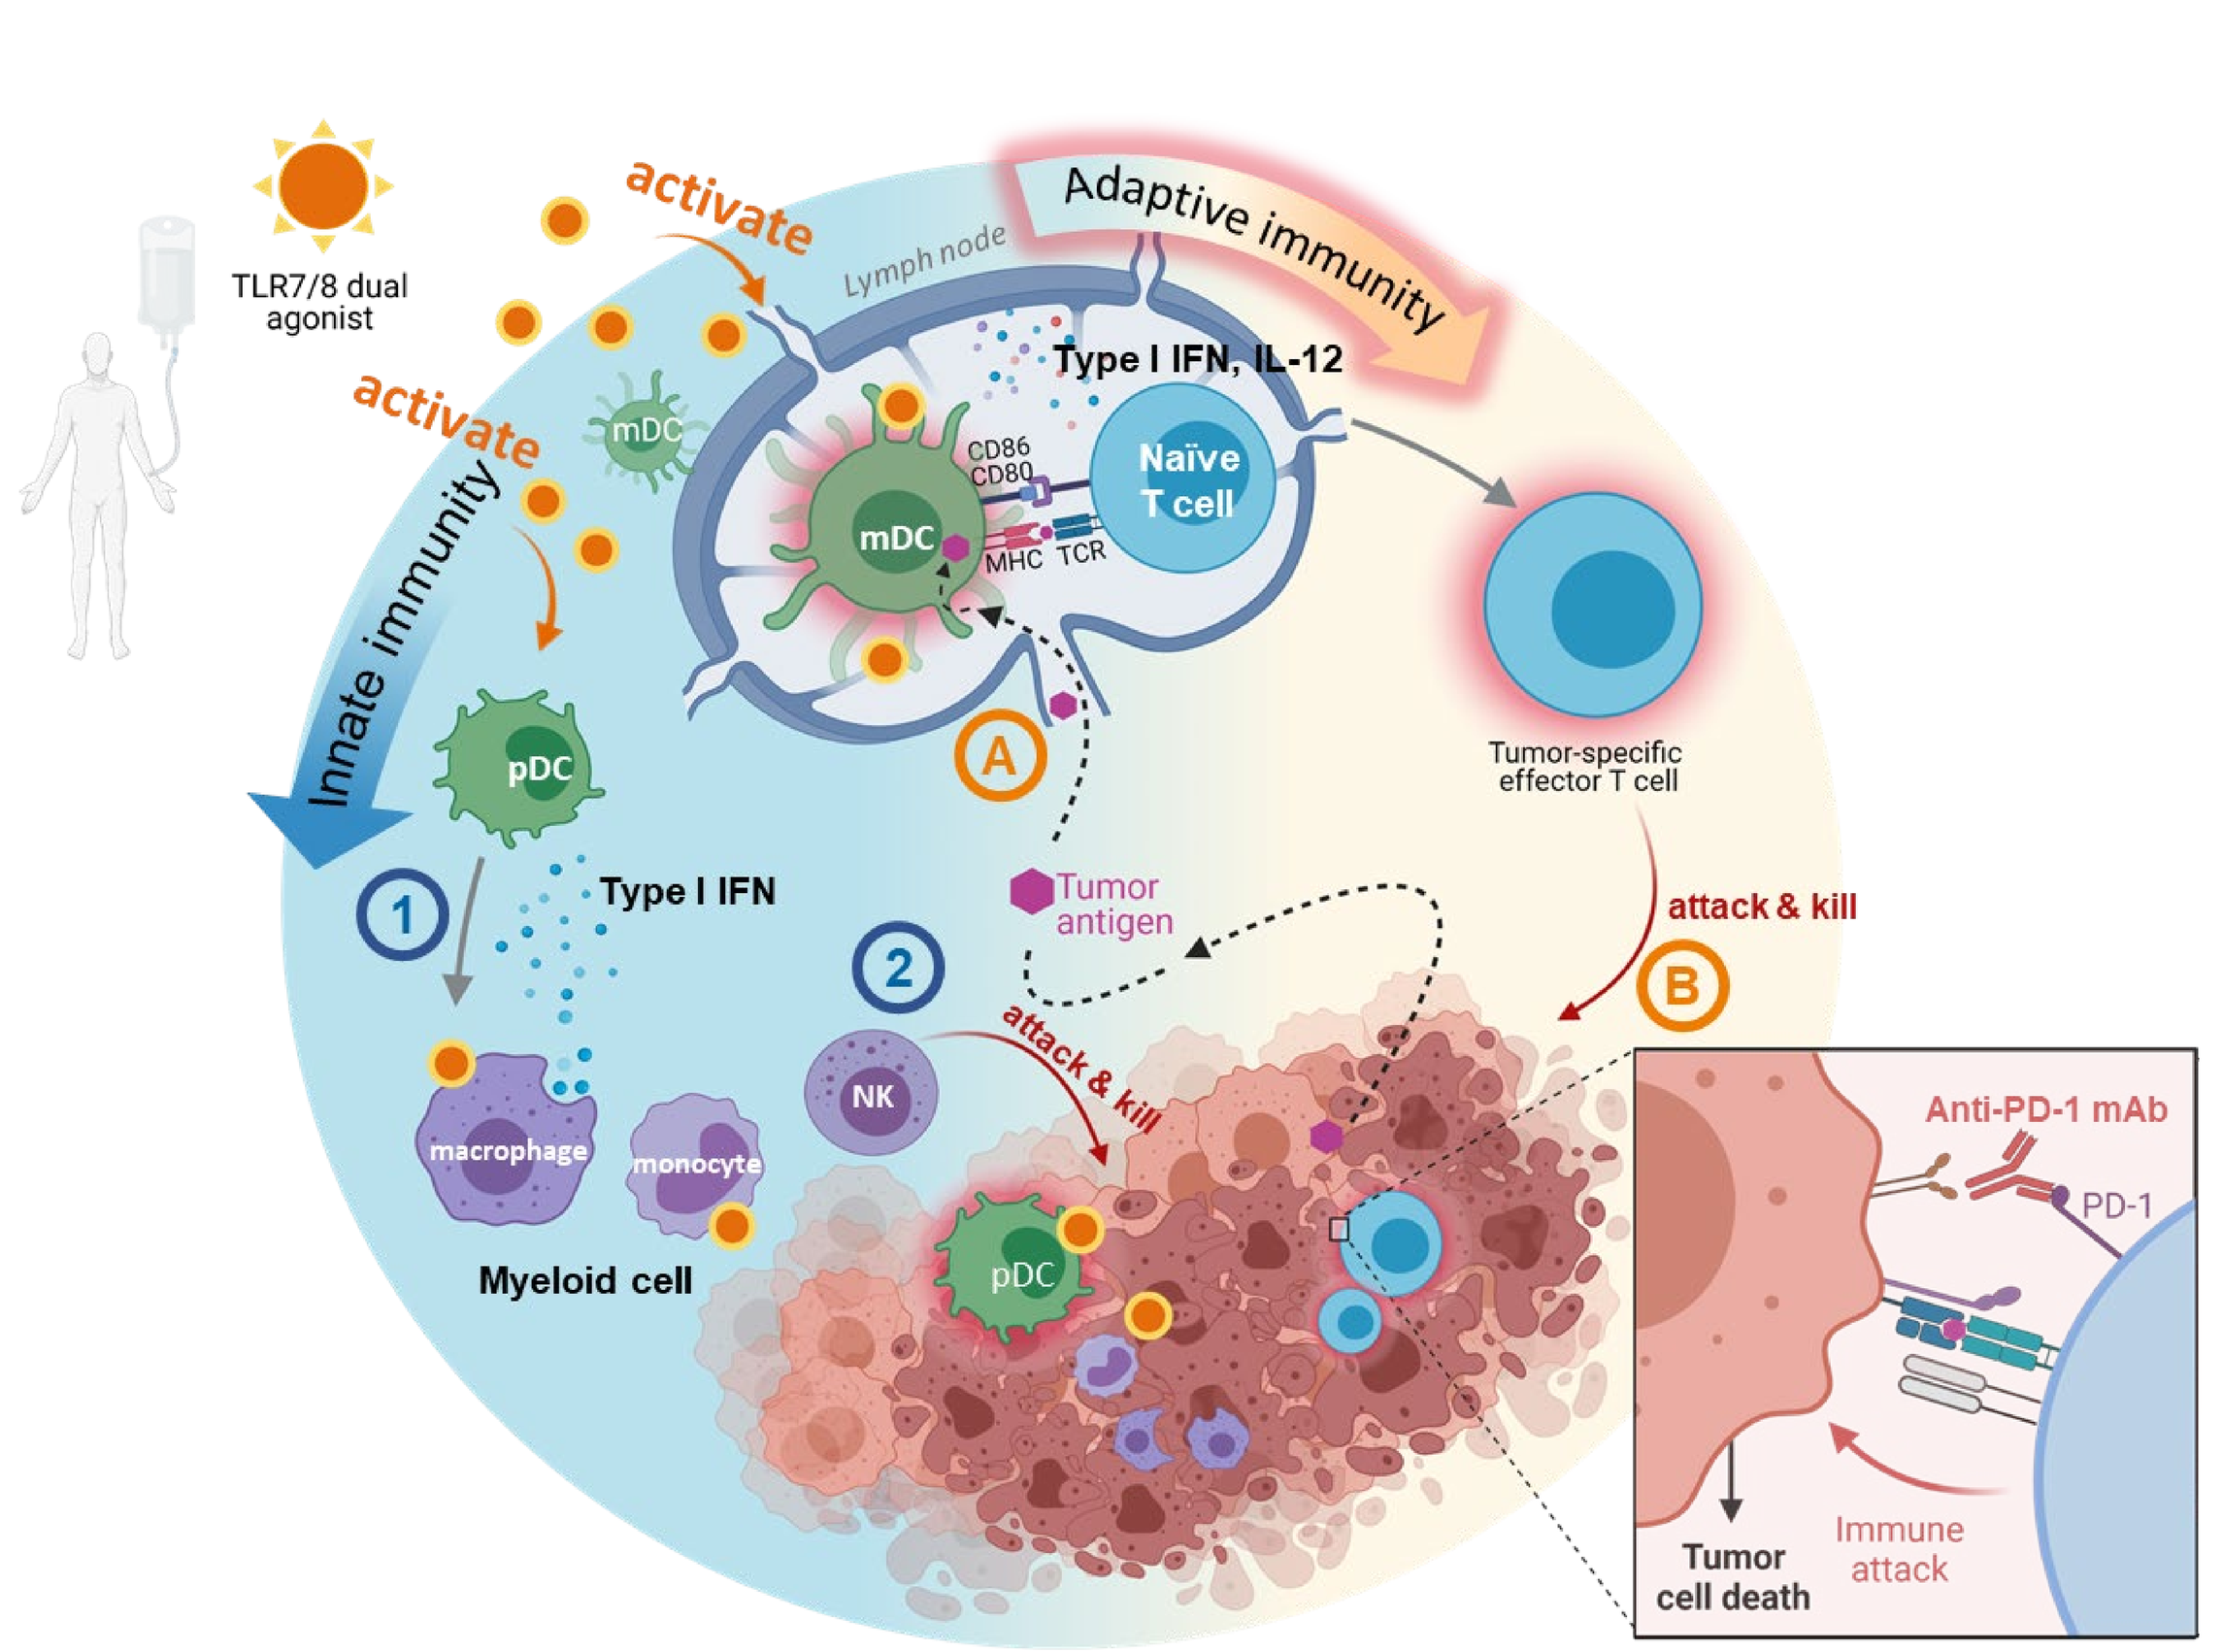

TLR7/8 dual agonist
activate
activate
Adaptive immunity
Lymph node
Innate immunity
Type I IFN, IL-12
mDC
CD86
CD80
MHC
TCR
Naïve T cell
Tumor-specific effector T cell
pDC
Type I IFN
Tumor antigen
attack & kill
attack & kill
NK
macrophage
monocyte
Myeloid cell
Anti-PD-1 mAb
PD-1
Tumor cell death
Immune attack

TLR7/8 dual agonist activates innate and adaptive immune responses

Innate immunity

TLR7 agonist induces pDC-driven innate immunity against tumor

1. Type I IFNs signals for NK cells activation
2. NK cells recruited to TME to kill tumors

Adaptive immunity

TLR8 agonist stimulates myeloid cells and mDCs which enhance tumor-specific T cell priming and activation

A. Activated mDCs take up, process and present tumor antigens to T cells
B. Tumor specific T cells expand and kill tumors. Effect enhanced by checkpoint inhibitors.

- ***Unlike most intratumoral TLR agonists in development, BDB001 is administered intravenously allowing for systemic activation of DCs and treatments of tumors more broadly***
- ***BDB001 monotherapy was previously reported to be well tolerated and elicit clinical responses (SITC, 2020)***

A multi-center, open-label, dose escalation / dose expansion Phase 1 study of BDB001 in combination with pembrolizumab

BDB001 in Combination w/ pembrolizumab (n=23)

Combo Dose Level 4
BDB001 Dose Level 4 IV Q1W + pembrolizumab IV Q3W

Combo Dose Level 3
BDB001 Dose Level 3 IV Q1W + pembrolizumab IV Q3W

Combo Dose Level 2
BDB001 Dose Level 2 IV Q1W + pembrolizumab IV Q3W

Combo Dose Level 1
BDB001 Dose Level 1 IV Q1W + pembrolizumab IV Q3W

Ongoing Combination Dose Expansion (n=14)

BDB018 MTD/RP2D + pembro

Combo Expansion Cohort
At least 20 additional patients with unresectable or metastatic solid tumors that have relapsed or are refractory to standard therapies or for whom there is no approved therapy

Primary Objective
- Safety

Secondary Objectives
- Efficacy
- Pharmacokinetics
- Pharmacodynamics

Data cut-off: 4/4/21

37 subjects evaluable for safety

28 subjects evaluable for efficacy
- **Dose Levels 3 and 4 (n=19)**

Clinical Trial ID: BDB001-101 / KN-914* (NCT03486301)
In collaboration with Merck & Co., Inc., Kenilworth, NJ, USA

Patient Characteristics	No.
Gender	
Female	23 (62%)
Male	14 (38%)
Age median, range	65 yrs (33-86)
Tumor types	13
Prior lines of systemic therapy (median, range)	3 (0-8)
Anti-PD-(L)1	15 (41%)
Chemotherapy	30 (81%)
Targeted therapy	28 (76%)

Tumor Characteristics	No.
PD-L1	
Positive	7 (19%)
Negative	11 (30%)
Unknown	19 (51%)
MSI	
High	2 (5%)
Stable	22 (59%)
Unknown	13 (35%)
TMB	
High	1 (3%)
Intermediate	2 (5%)
Low	7 (19%)
Unknown	27 (73%)

Safety and Efficacy Results

BDB001 combined with pembrolizumab was well tolerated without a DLT with a favorable efficacy profile

Safety Profile

Treatment related AEs occurring in > 5% of subjects or ≥ Grade 3, (N=37)

Adverse events (AEs, CTCAE v. 4.03)	BDB001 + pembrolizumab All grades	BDB001 + pembrolizumab Grade 3
All AEs	29 (78.4)	
Chills / Rigors	12 (32.4)	
Fatigue	12 (32.4)	2 (5.4)
Fever	12 (32.4)	
Pruritus / Rash	5 (13.5)	1 (2.7)
Infusion Related Reaction	4 (10.8)	
Hypothyroidism	4 (10.8)	
AST elevation	3 (8.1)	
Diarrhea	3 (8.1)	
Nausea	3 (8.1)	
ALT elevation	2 (5.4)	
Anorexia	2 (5.4)	
Blood bilirubin increase	2 (5.4)	
Cytokine Release Syndrome (CRS)	2 (5.4)	
Vomiting	2 (5.4)	
ALP elevation	1 (2.7)	1 (2.7)
Mucositis or stomatitis (oral)	1 (2.7)	1 (2.7)
Muscle weakness	1 (2.7)	1 (2.7)
Pancreatitis	1 (2.7)	1 (2.7)

Efficacy Profile

Best Overall Response	BDB001 + pembrolizumab Dose Level 3 and 4 Subjects (%), (n=19)
Complete response (CR)	**1 (5)***
Partial response (PR)	**4 (21)***
Stable disease (SD)	6 (32)†
Progressive disease (PD)	8 (42)
Disease control rate (CR+PR+SD)	11 (58)
Overall response rate (CR+PR)	**5 (26)**

Note: 19 subjects were evaluable for efficacy treated with either BDB001 at Dose Level 3 or 4 across Dose Escalation and Expansion phase

* Responses in cholangiocarcinoma, HCC, melanoma, ovarian carcinoma and breast cancer – triple negative; Unconfirmed PR in HCC subject

† 4 out 6 SD patients had reduced tumor burden and prolonged stable diseases >18 weeks; near PR in cholangiocarcinoma subject (active, 29% reduction in tumor burden)

Note: Safety data as of 4/4/2021

- Overall, BDB001 combined with pembrolizumab was well tolerated
- Over 21% of the subject did not experience any treatment-related AE's (TRAE's) and there were no Grade 4 or 5 TRAE's
- The combination treatment did not substantially increase Grade 3 or 4 TRAE's compared to BDB001 monotherapy
- Most common TRAE's were immune-related (fever, chills, and rigors) and fatigue
- Clinical responses observed in patients treated with BDB001 at Dose Levels 3 and 4 in combination with pembrolizumab
- Based on clinical and PK-PD data, BDB001 Dose Levels 3 in combination with pembrolizumab was selected for Dose Expansion

BDB001 combined with pembrolizumab leads to deep, durable clinical responses including subjects with anti-PD-(L)1 refractory tumors

Responses in evaluable subjects treated with BDB001 RP2D (Dose Level 3 or 4) + pembrolizumab (N=19)

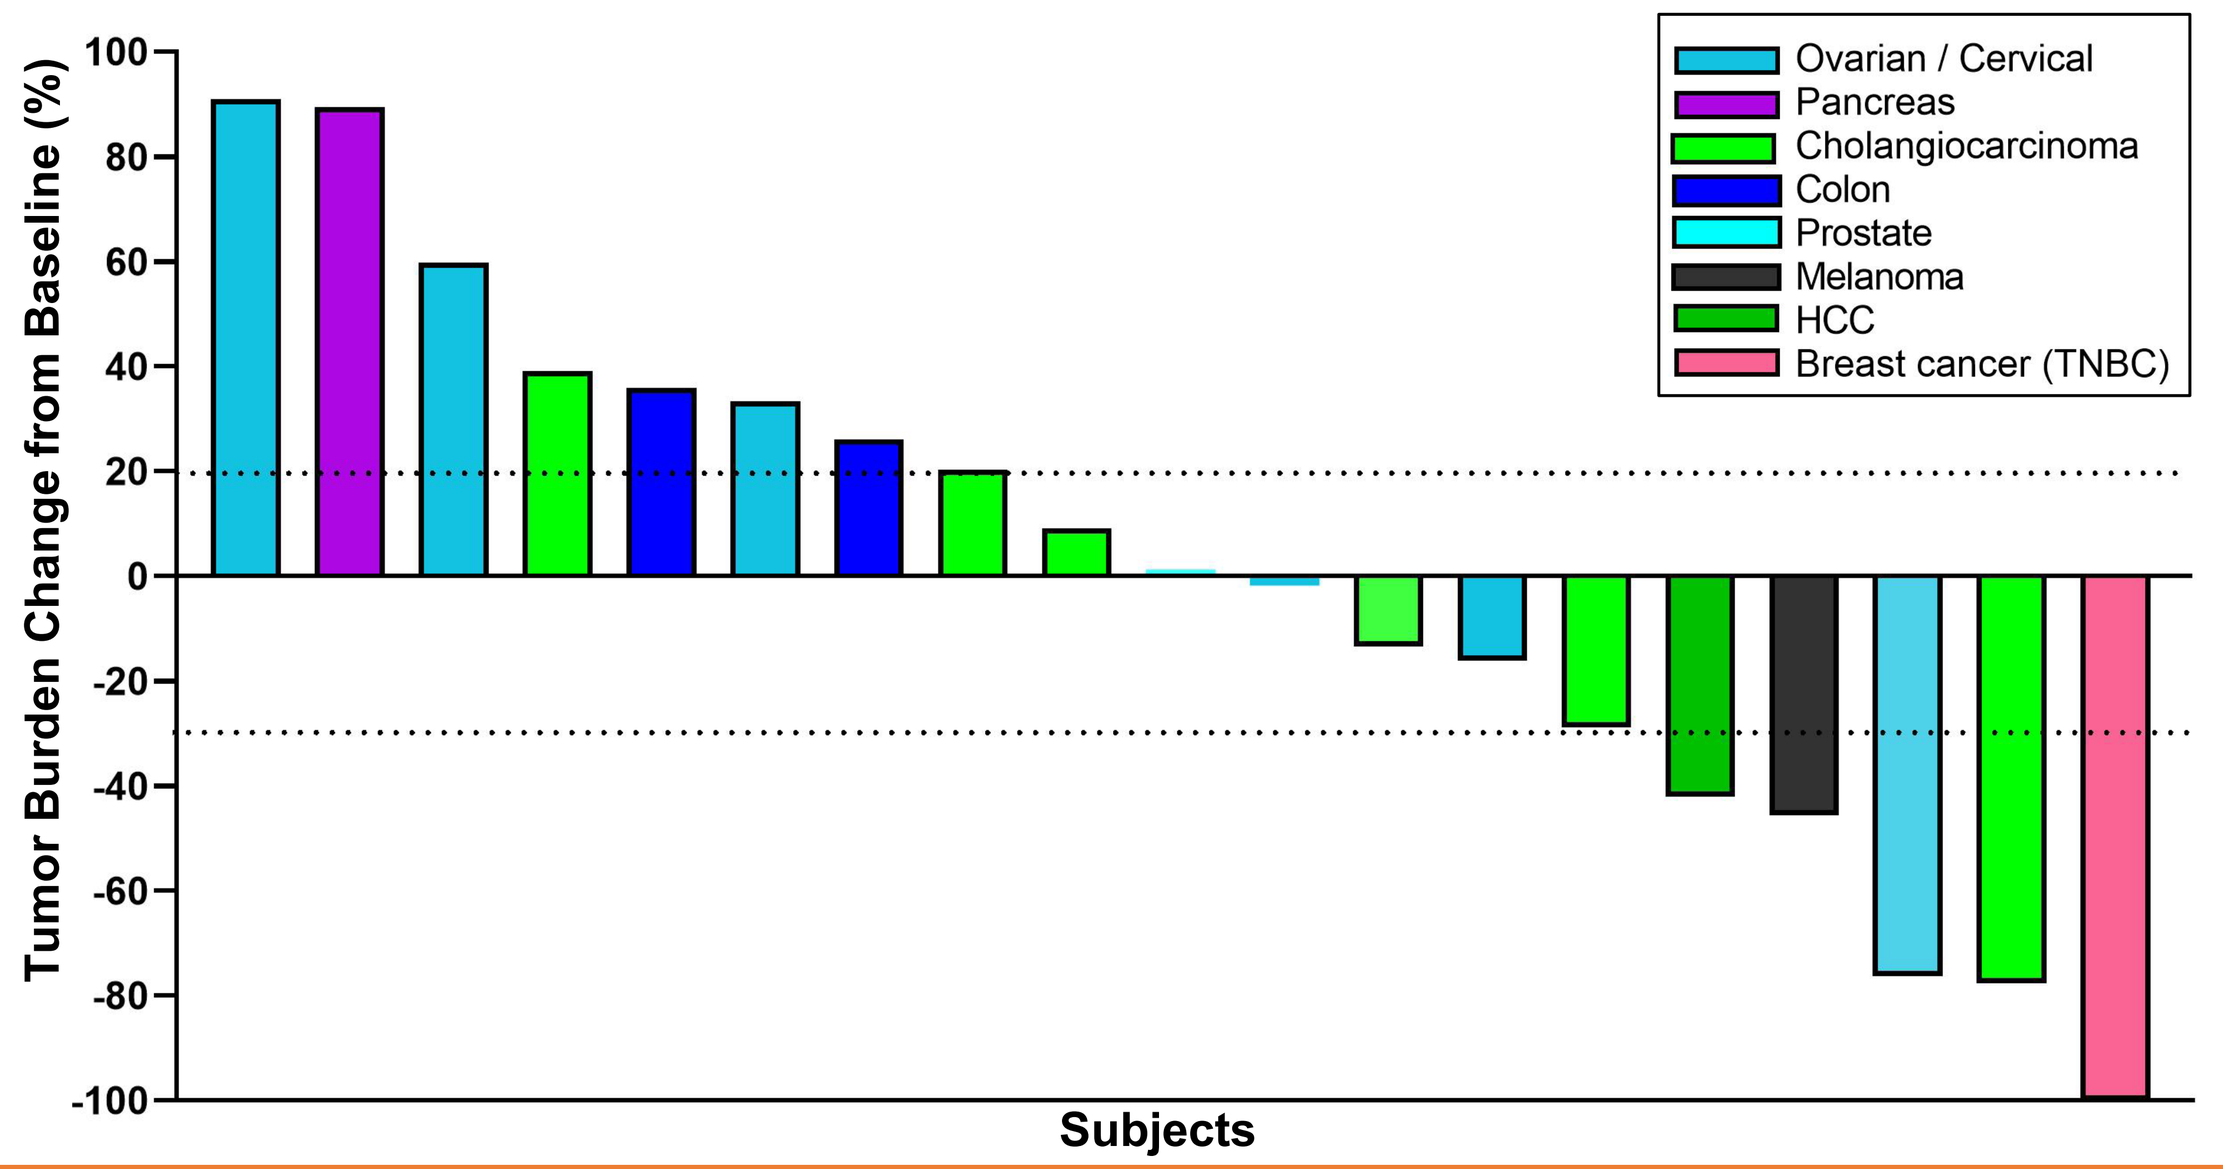

Tumor Burden Change from Baseline (%)
100
80
60
40
20
0
-20
-40
-60
-80
-100
Subjects
Ovarian / Cervical
Pancreas
Cholangiocarcinoma
Colon
Prostate
Melanoma
HCC
Breast cancer (TNBC)

Efficacy Endpoints

- Overall response rate
 - RECIST 1.1 / irRECIST: **26%**
- Time to response (median, range)
 - **8.6 weeks** [4.3 – 18.4 weeks]
- On study for more than 18 weeks (≥2 efficacy assessments)
 - **4** responders (one of whom on treatment >47 weeks / 5 efficacy assessments)
 - **1** more responder has next efficacy assessment at 18 weeks

Clinical responses in tumors with low response rate to anti-PD-1 therapy*

Tumor type	BOR BDB001+ pembro	Prior therapies	PD-L1 status	MSI status	Tumor Mutational Burden
Breast cancer – triple negative¶	CR	• 3 lines of chemotherapy – platinum-resistant • 1 line of antibody-drug conjugate	Negative	Stable	Low
Cholangiocarcinoma¶	PR	• 3 lines of chemotherapy	Negative	Stable	Low
Ovarian carcinoma¶	PR	• 4 lines of chemotherapy +/- anti-angiogenesis agent – platinum-resistant	Negative	Unknown	Unknown
Melanoma	PR	• 2 lines of Immunotherapy including anti-PD-1 (pembrolizumab)†	Unknown	Unknown	Unknown
Hepatocellular carcinoma	PR	• 2 lines of anti-angiogenesis agent • 1 line of immunotherapy, including anti-PD-L1 (durvalumab)†	Unknown	Stable	Unknown
Cholangiocarcinoma	SD (near PR)	• 3 lines of chemotherapy	Unknown	Stable	Low

* The probability of response to anti-PD-1 monotherapy is < 5% in subjects responded to BDB001+pembrolizumab; ¶See panel to the right for scan images; †Anti-PD-(L)1 therapies were NOT immediately prior to BDB001+pembro

Efficacy and Pharmacodynamic Results

Clinical responses observed in subjects with various tumor types

54-year-old Female
Metastatic Breast Cancer
Triple Negative
Complete Response

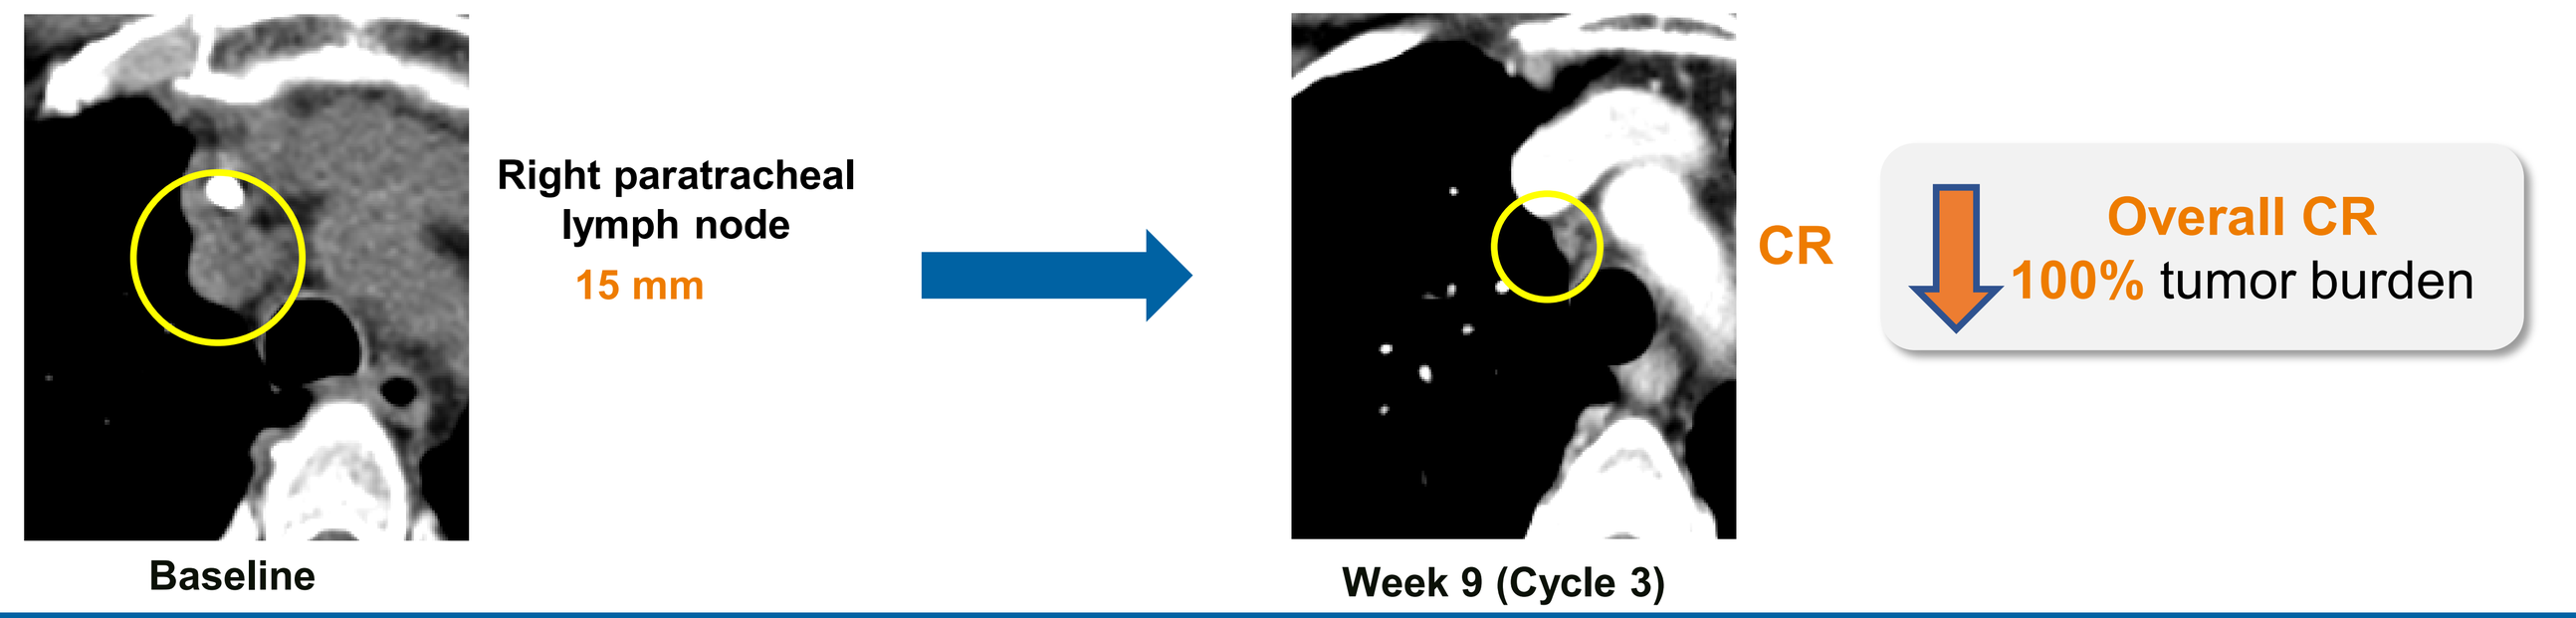

Right paratracheal lymph node
15 mm
CR
Overall CR
100% tumor burden
Baseline
Week 9 (Cycle 3)

56-year-old Female
Metastatic
Cholangiocarcinoma
Partial Response

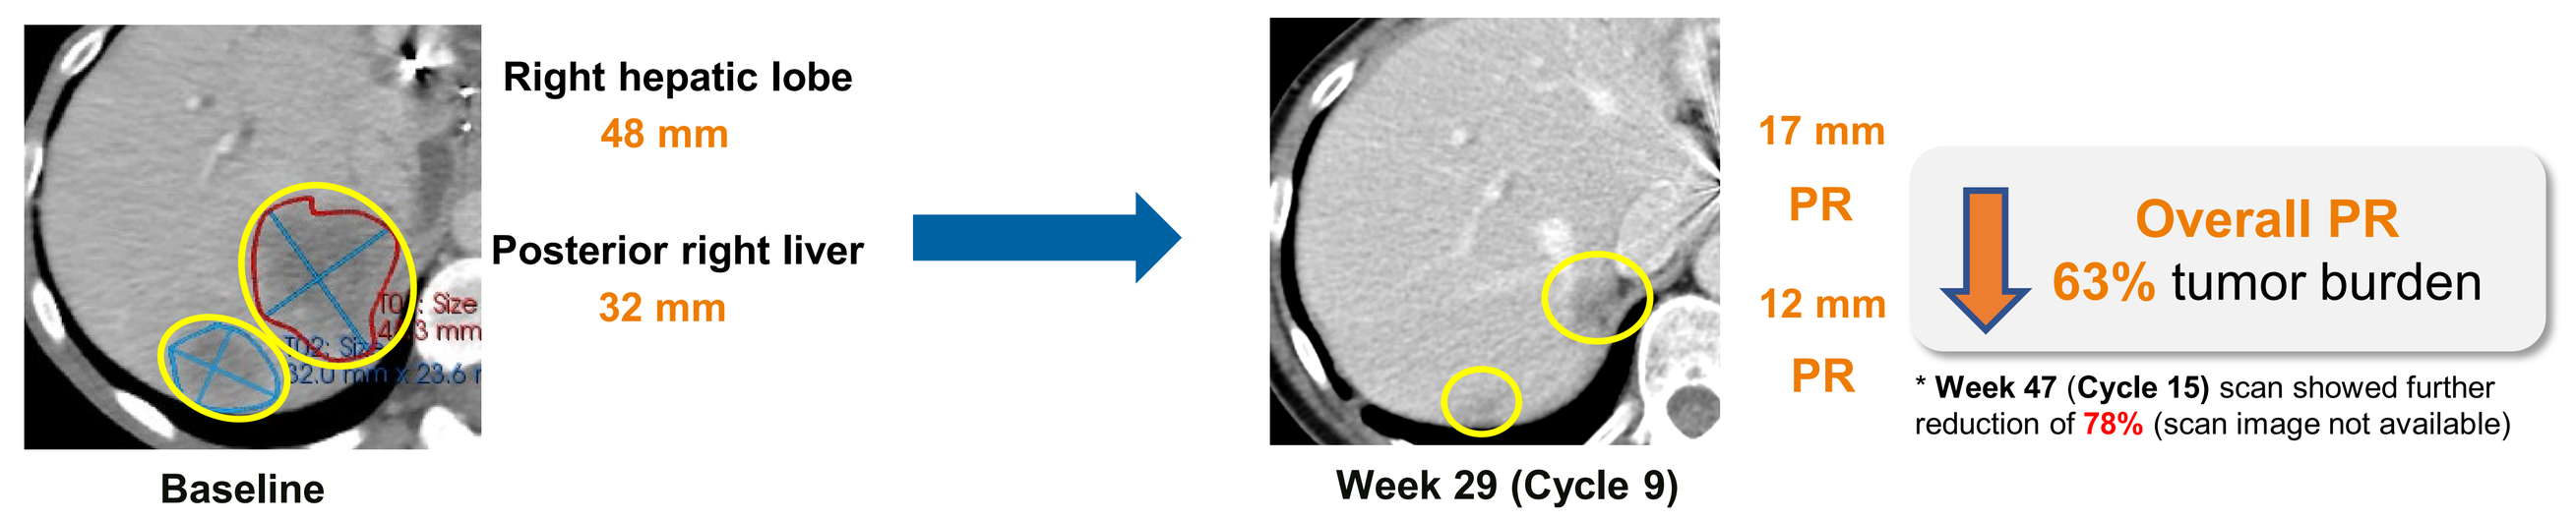

Right hepatic lobe
48 mm
Posterior right liver
32 mm
17 mm
PR
12 mm
PR
Overall PR
63% tumor burden
* Week 47 (Cycle 15) scan showed further reduction of 78% (scan image not available)
Baseline
Week 29 (Cycle 9)

46-year-old Female
Metastatic Ovarian Cancer
(Platinum Resistant)
Partial Response

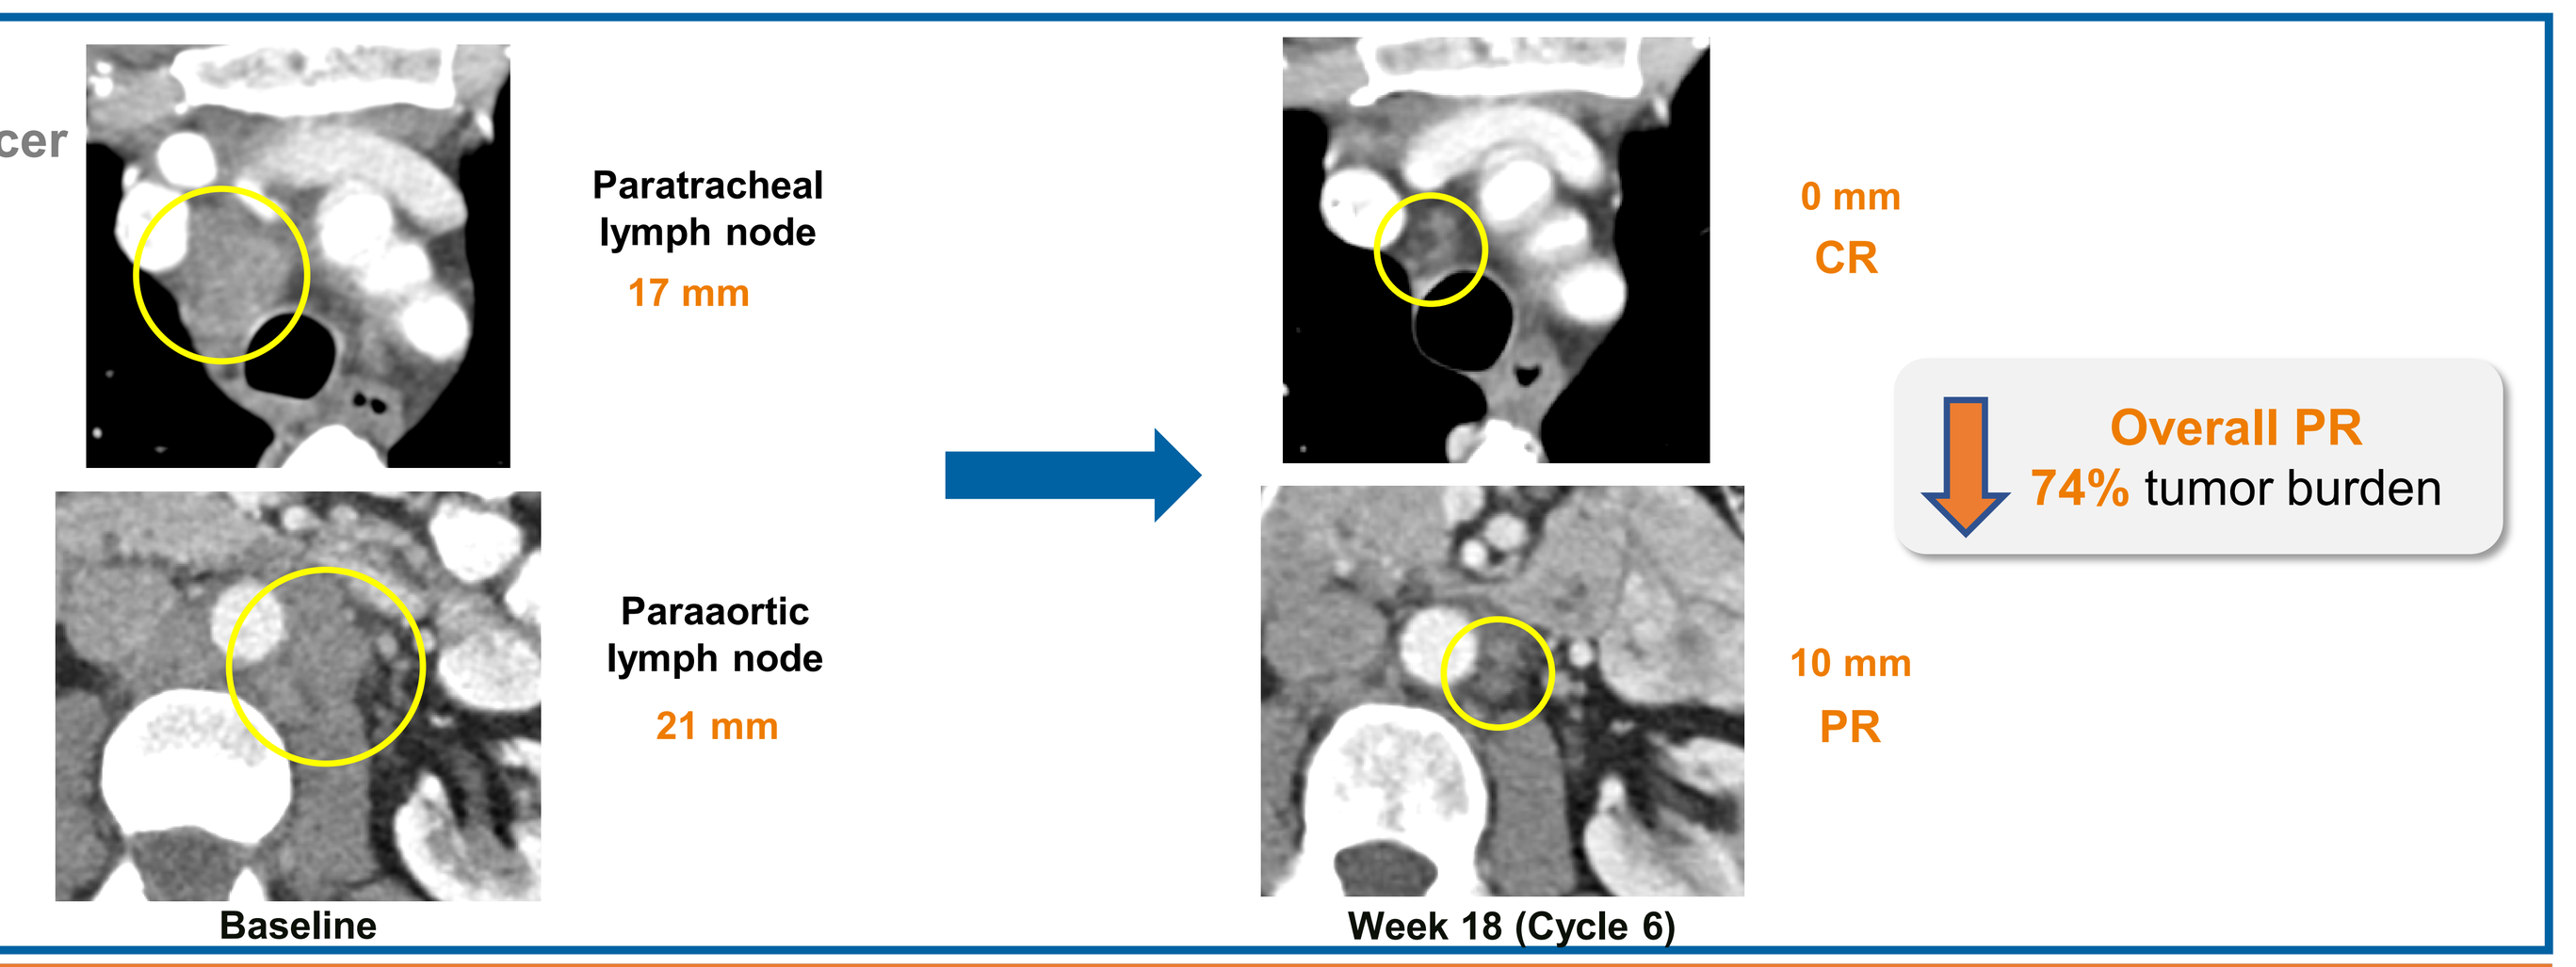

Paratracheal lymph node
17 mm
0 mm
CR
Overall PR
74% tumor burden
Paraaortic lymph node
21 mm
10 mm
PR
Baseline
Week 18 (Cycle 6)

BDB001+pembro activates anti-tumor immunity systemically

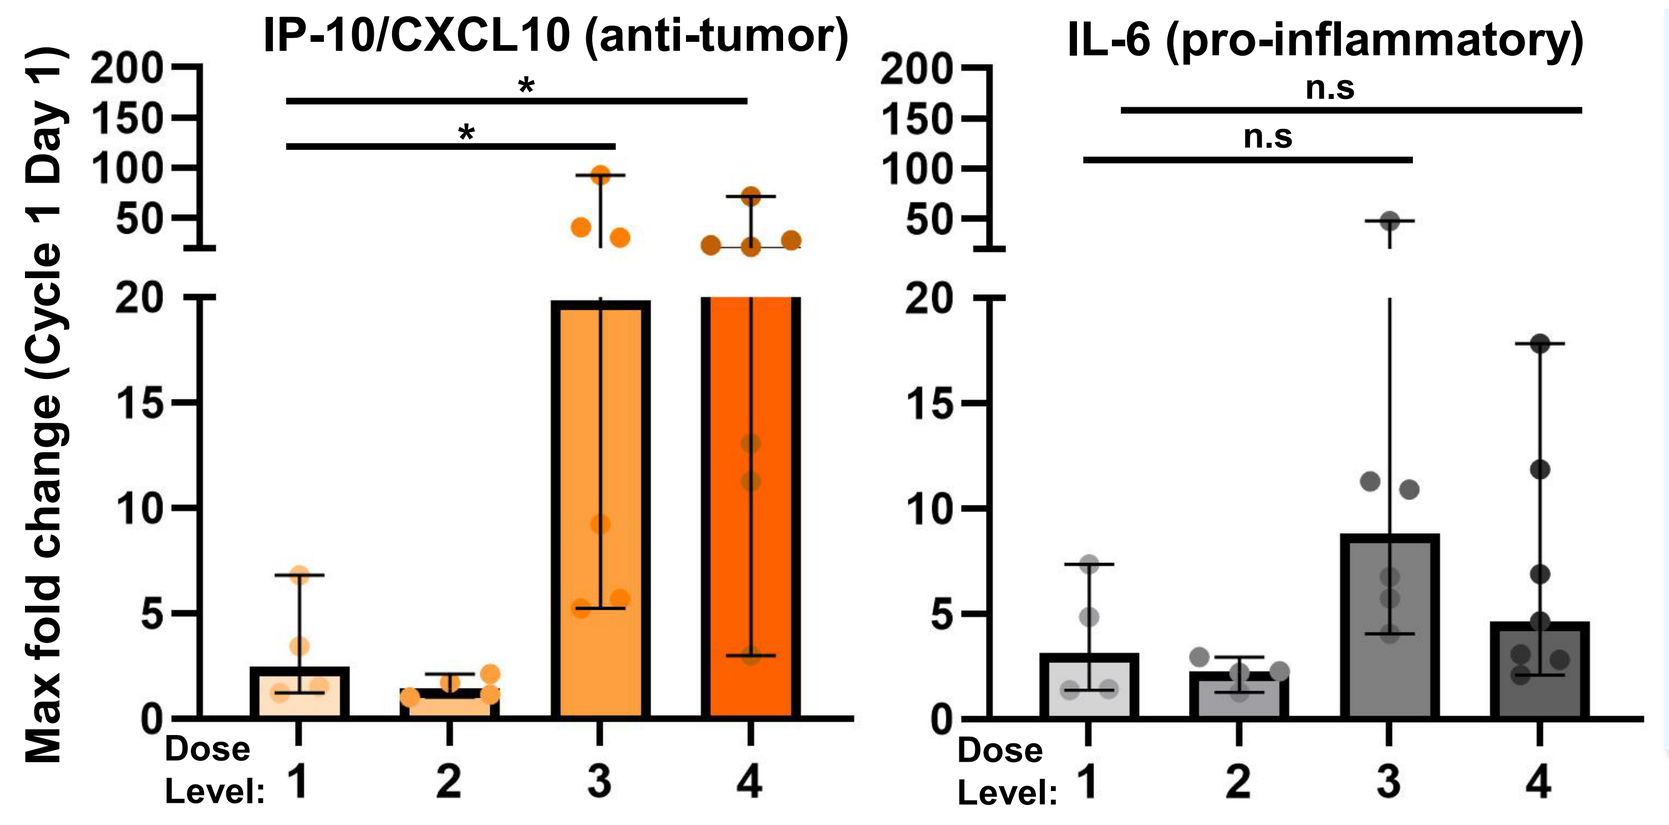

IP-10/CXCL10 (anti-tumor)
IL-6 (pro-inflammatory)
Max fold change (Cycle 1 Day 1)
*
*
n.s
n.s
Dose Level: 1 2 3 4

- BDB001 dose-dependent increase in plasma IP-10 levels (statistically significant), indicative of anti-tumor immune activation
- IP-10 induction by BDB001 combined with pembrolizumab associated with clinical responses
- No significant increase in inflammatory cytokine IL-6 levels
- Overall, BDB001+pembro favors induction of anti-tumor cytokine IP-10 over pro-inflammatory/CRS markers, IL-6, IL-10, TNF-α, and MCP-1

*Plasma cytokine levels; *P < 0.05 (Mann-Whitney Test); n.s = statistically not significant*

Discussion / Conclusions

- Seven and Eight Biopharma's systemic delivery of TLR 7/8 dual agonist **BDB001 is first-in-class**
- **BDB001 was delivered safely intravenously in combination with pembrolizumab**
- ≤ Grade 2 (no Grade 3/4) CRS or IRR, as deemed by the investigators, were seen in 6 subjects (16%). In these subjects:
 - AE's occurred hours after the first infusion of BDB001 and pembrolizumab and all symptoms resolved quickly without long-term sequalae
 - None led to infusion stoppage or study drug discontinuation
 - None required hospitalization or vasopressors
 - Symptoms included fever, chills, and rigors, similar to AE's commonly seen in subjects deemed not to have CRS or IRR
 - IL-6 levels and other CRS markers were not elevated in the subjects deemed to have CRS or IRR
 - IL-6 levels were >100 times lower than levels seen in subjects with CAR-T-induced CRS
- BDB001 in combination with pembrolizumab had robust dose dependent immune activation without increased risk of CRS
- **BDB001 combined with pembrolizumab resulted in clinical responses** in tumors that had progressed on multiple prior therapies including anti-PD-(L)1 therapy and with low probability of responding to anti-PD-1 therapy based on their PD-L1 negative, MSI-stable, and Tumor Mutational Burden-low status
- Based on safety, efficacy, and PK-PD results, BDB001 Dose Level 3 combined with pembrolizumab was selected for the ongoing Dose Expansion part of the trial to further evaluate safety, efficacy, and immune modulatory effects in the tumor microenvironment
- **BDB001 in combination with pembrolizumab represents a novel and viable treatment for advanced solid tumors**

Presented at ASCO 2021 (Virtual)

Copyright © 2021 **Seven and Eight Biopharmaceuticals Inc.** All rights reserved.